Preclinical Pharmacokinetic and Safety Profile of IDX375, A Novel and Potent Non-Nucleoside HCV Polymerase Inhibitor

[1]S.S. Good, [1]C. Bu, [1]M. Camire, [1]B. Hernandez-Santiago, [1]M. Larsson, [1]H. Rashidzadeh, [1]X.-R. Pan-Zhou, [1]S. Luo, [1]J. Selden, [2]C.B. Dousson and [2]D. Surleraux
[1]Idenix Pharmaceuticals, Inc., Cambridge, MA, USA; [2]Idenix Pharmaceuticals, Inc., Montpellier, France

Email: good.steven@idenix.com

#323

INTRODUCTION

- Hepatitis C virus (HCV) is a common blood-borne pathogen annually infecting three to four million people worldwide. Currently, an estimated 170 million people are infected globally.[1]
- The current standard-of-care therapy, a combination of pegylated interferon and ribavirin, is effective in only 50% of patients infected with genotype 1 HCV and is associated with significant side effects. Thus, there remains a need for new, more effective and better tolerated HCV treatment options.[2,3]
- The HCV polymerase is an attractive antiviral target. Nucleoside analogs or, more recently, pro-nucleotides such as IDX184, which target the active site of the enzyme, are currently in clinical trials. In another approach, multiple classes of non-nucleoside polymerase inhibitors (NNI), which target different allosteric sites of the enzyme, are under investigation.
- IDX375 is a novel NNI clinical candidate that targets the palm pocket of the NS5B polymerase. This study evaluated the preclinical pharmacokinetics, *in vitro* metabolism and preliminary toxicology profiles of IDX375.

METHODS

PK methods: Male mice (3/time point), rats (3/dose group), dogs and monkeys (2/dose group) were given a single IV or PO dose of IDX375. Unchanged drug was quantified in samples of plasma and liver (rodents only) by HPLC-MS/MS using a protein precipitation or a liquid-liquid extraction method. PK parameters were calculated using Kinetica or WinNonlin.

***In vitro* metabolism assays:** Freshly isolated hepatocytes were incubated with ^{14}C-IDX375 (1 or 5 µM) for up to 4 h. Reactions were terminated with acetonitrile and the disappearance of IDX375 was determined by HPLC using a Radiomatic flow detector.

***In vitro* cytotoxicity assays:** Freshly isolated hepatocytes were incubated with various concentrations of IDX375 for 48 h. Intracellular ATP content was measured (CellTiter-Glo luminescent cell viability assay) to determine cell cytotoxicity (CC_{50}).

CYP inhibition assays: IDX375 was incubated with human CYP450 isozymes and appropriate fluorometric substrates according to the protocol (BD Bioscience). For CYP2C9, a luminogenic substrate was used with the P450-Glo™ kit.

Preliminary monkey toxicology study: Cynomolgus monkeys (1/sex/group) were given daily oral doses of IDX375 (10 or 100 mg/kg/day) for seven days. Serial plasma samples were collected on days 1 and 7 and a portion of each liver was collected at necropsy on day 8 (~29 h post-dose). IDX375 plasma and liver concentrations were determined as described herein. Toxicity was evaluated based on mortality, clinical signs, body weight, qualitative food consumption, hematology, coagulation, clinical chemistry and urinalysis. Necropsy procedures consisted of gross pathology and organ weight measurements. Microscopic examinations were performed on all tissues collected from all animals.

RESULTS

Favorable Pharmacokinetics in Mice, Rats, Dogs and Monkeys

The pharmacokinetics of IDX375 was studied in mice and rats given a 15 mg/kg IV or PO dose, and in dogs and monkeys given a 2 mg/kg IV or a 10 mg/kg PO dose.

Figure 1: IDX375 PK Profiles in the Mouse

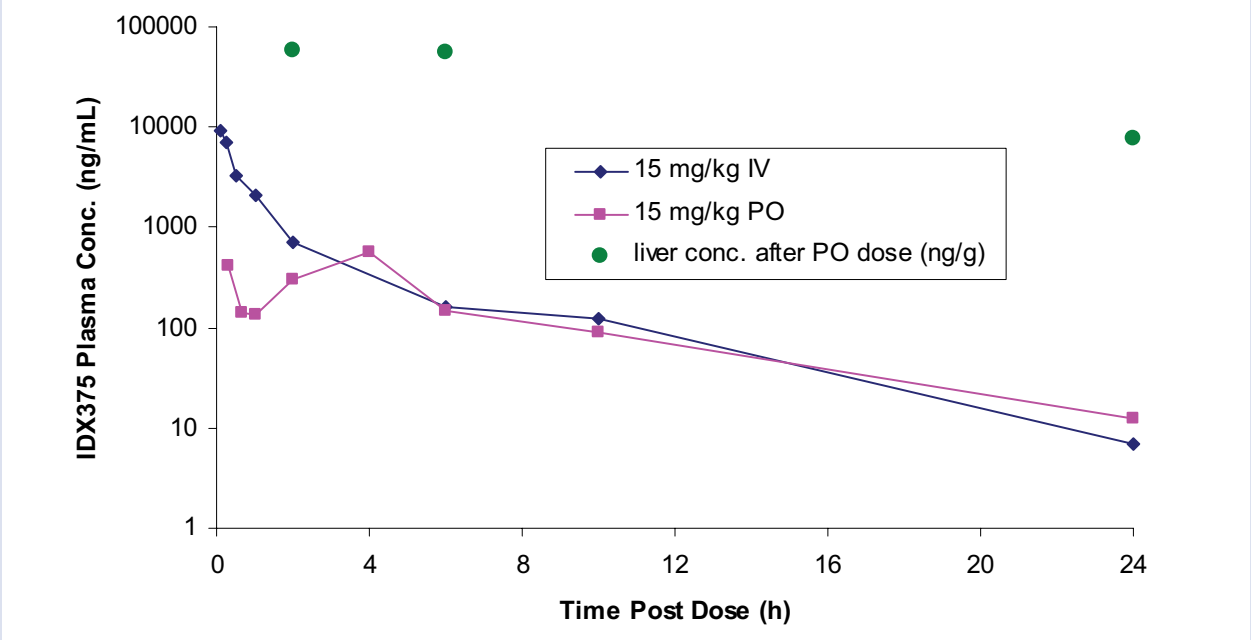
IDX375 Plasma Conc. (ng/mL)
Time Post Dose (h)
15 mg/kg IV
15 mg/kg PO
liver conc. after PO dose (ng/g)

Figure 2: IDX375 PK Profiles in the Rat

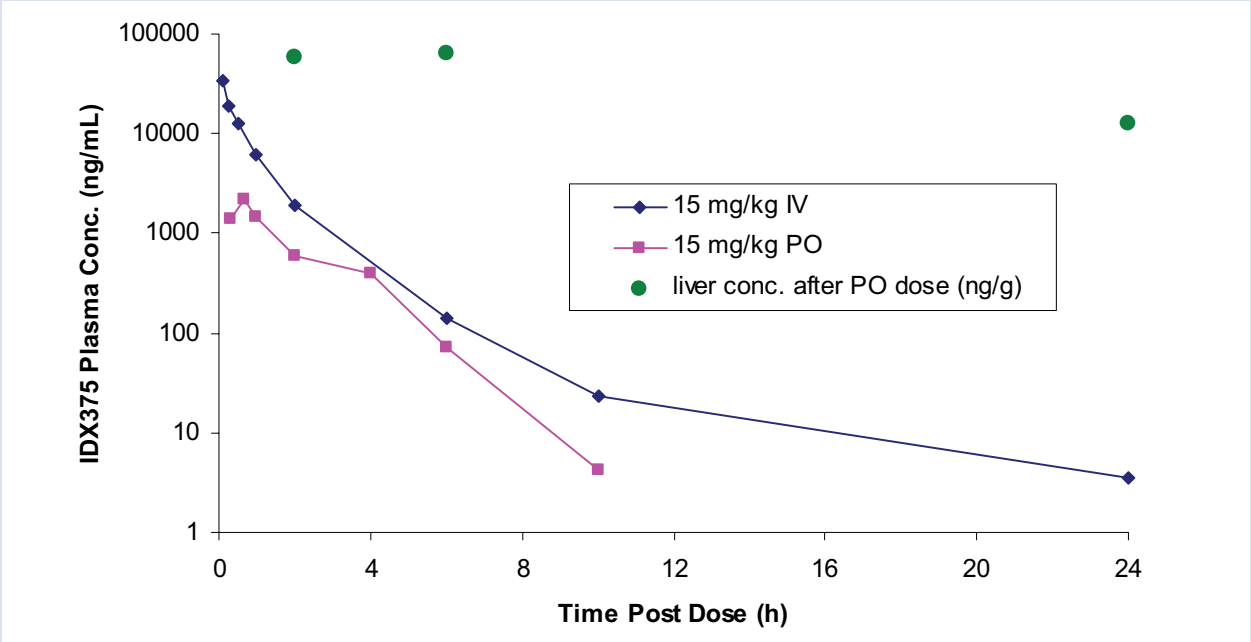
IDX375 Plasma Conc. (ng/mL)
Time Post Dose (h)
15 mg/kg IV
15 mg/kg PO
liver conc. after PO dose (ng/g)

Figure 3: IDX375 PK Profiles in the Dog

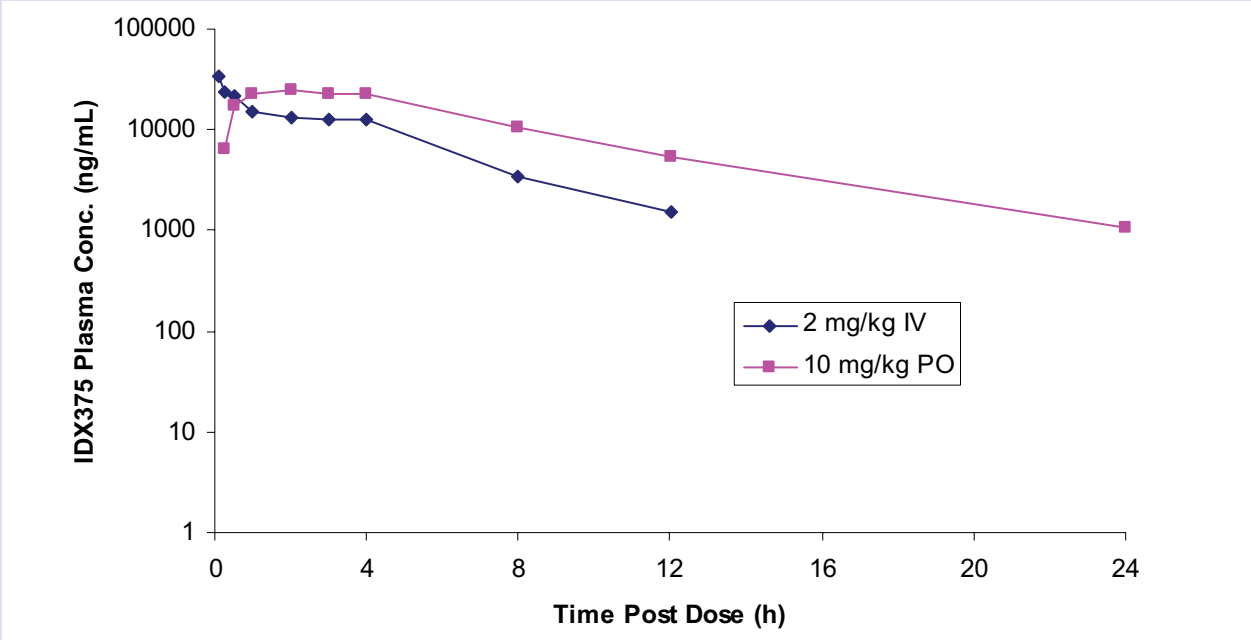
IDX375 Plasma Conc. (ng/mL)
Time Post Dose (h)
2 mg/kg IV
10 mg/kg PO

Figure 4: IDX375 PK Profiles in the Monkey

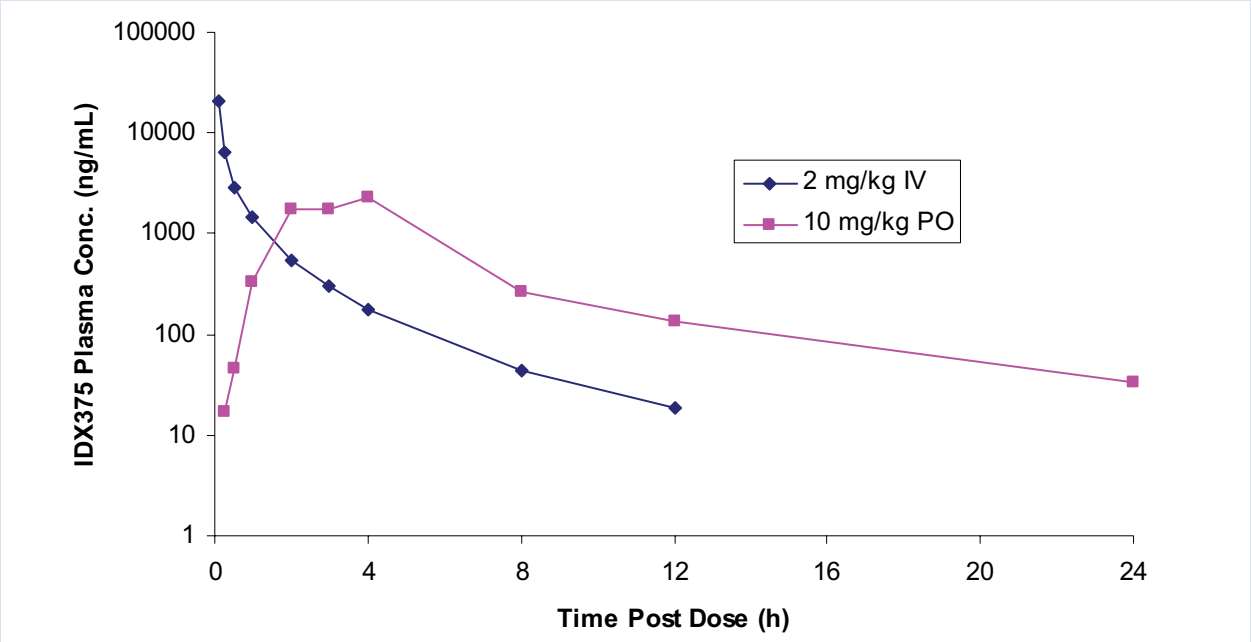
IDX375 Plasma Conc. (ng/mL)
Time Post Dose (h)
2 mg/kg IV
10 mg/kg PO

Table 1: IDX375 Preclinical Plasma PK Parameters

	Dose (mg/kg)	CL (L/h/kg)	V_d (L/kg)	C_{max} (ng/mL)	T_{max} (h)	$T_{1/2}$ (h)	F
mouse	15 (IV) 15 (PO)	1.6	9.9	569	4.0	5.1	34%
rat	15 (IV) 15(PO)	0.61	1.47	2210	0.67	1.4	16%
dog	2 (IV) 10 (PO)	0.02	0.08	24500	2.0-3.0	4.9	42%
monkey	2 (IV) 10 (PO)	0.24	0.77	2250	4.0	5.5	28%

CL and V_d are from IV data; C_{max}, T_{max} and $T_{1/2}$ are from PO data

- IDX375 demonstrated good oral bioavailability in all species studied.
- IDX375 was selectively concentrated in the liver; at 2, 6 and 24 h after a 15 mg/kg oral dose, liver levels in mice **(Figure 1)** and rats **(Figure 2)** were 60- to 700-times the corresponding plasma levels. In two monkeys given 10 mg/kg daily oral doses for 7 days, liver levels 29 h after the last dose were 54- and 59-fold higher than the corresponding 24-h plasma levels.

Favorable Exposures in Mice and Monkeys at High Oral Doses

IDX375 exposure ($AUC_{0\text{-}24h}$) was determined in mice and rats given single doses up to 250 mg/kg and in monkeys given daily doses up to 100 mg/kg for 7 days.

Figure 5: IDX375 Exposure-Dose Response Curves

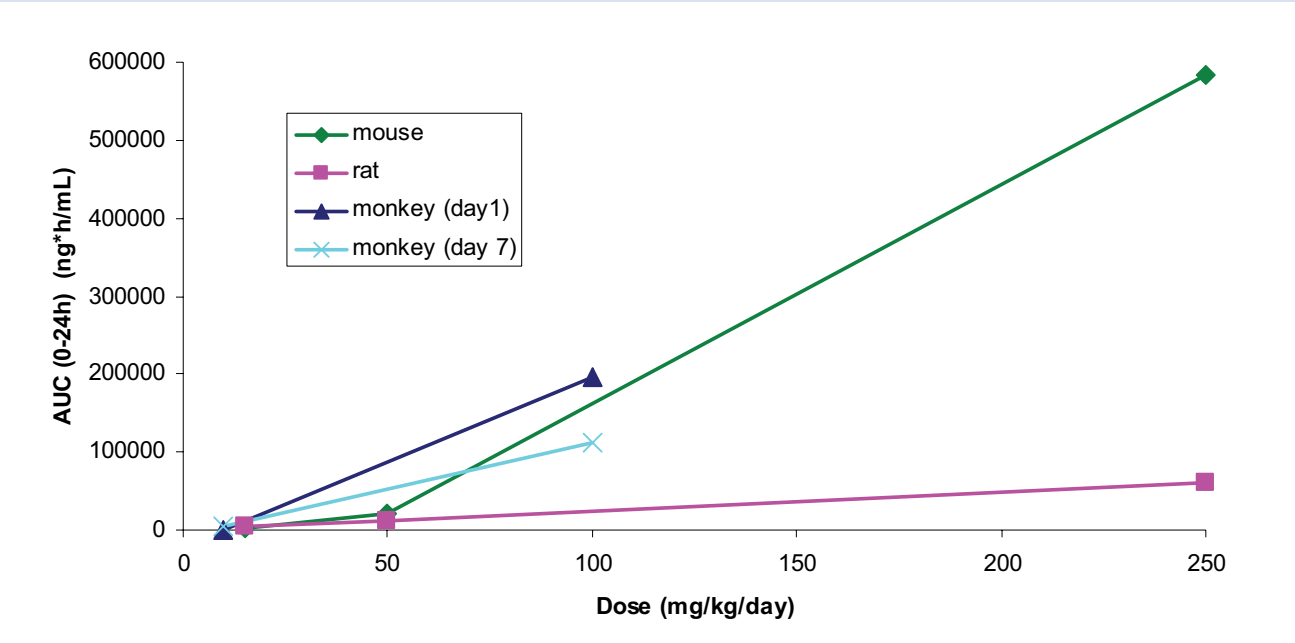
AUC (0-24h) (ng*h/mL)
Dose (mg/kg/day)
mouse
rat
monkey (day1)
monkey (day 7)

- In the dose ranges studied, $AUC_{0\text{-}24h}$ values increased in a dose-proportional manner in rats and were greater than dose-proportional in mice and monkeys.

Limited Metabolism and No Cytotoxicity in Hepatocytes

The metabolism and cytotoxicity of IDX375 were studied in freshly isolated mouse, rat, monkey and human hepatocytes.

Table 2: Cytotoxicity and Half-Life of IDX375 in Hepatocytes

Primary Hepatocytes	CC_{50} (µM)	Mean Half-Life (h) ± SD*	
		1 µM	5 µM
mouse	>10	6.1 ± 1.3	10.8 (2)
rat	>10	6.8 ± 2.4	7.6 ± 2.7
monkey	>10	3.1 ± 1.9	4.4 ± 3.1
human	>10	6.1 ± 3.6	7.7 (2)

*n=3 except where indicated

- IDX375 was not cytotoxic to mouse, rat, monkey or human hepatocytes.
- The extent of metabolism of IDX375 was greatest in monkey hepatocytes; half-lives were similar in mouse, rat and human hepatocytes.

Limited Inhibition of Human CYP450 Isozymes

The potential of IDX375 to inhibit human CYP450 enzymes was studied using supersomes expressing specific isozymes.

Table 3: Inhibition of Human CYP450 by IDX375

Human CYP450	Probe Substrate	IC_{50} (µM)
1A2	3-cyano-7-ethoxycoumarin	no inhibition
2B6	7-ethoxy-4-trifluoromethylcoumarin	>20
2C9	Luciferin-H	>20
2D6	3-[2-(N,N-diethyl-N-methylamino)-methyl]-7-methoxy-4-methylcoumarin	>20
3A4	7-benzyl-trifluoromethylcoumarin	>20

20 µM was the highest concentration tested

- IDX375 did not significantly inhibit any of the five major human CYP450 isozymes tested.

No Adverse Effects Observed in a Monkey Toxicology Study

Monkeys were given single daily oral doses of 10 or 100 mg/kg IDX375 for 7 days.

- Substantial plasma exposures were achieved in monkeys; increasing the dose from 10 to 100 mg/kg resulted in a 22-fold increase in the mean day-7 $AUC_{0\text{-}24h}$ values **(Figure 5)**.
- No meaningful changes in clinical chemistry parameters were detected.
- No histological abnormalities were observed.

CONCLUSIONS

- The favorable preclinical pharmacokinetics of IDX375 suggests the potential for once- or twice-daily oral dosing in HCV-infected patients.
- *In vitro* data showed limited metabolism in human hepatocytes and minimal inhibition of human CYP450s.
- IDX375 demonstrated no adverse effects in a preliminary toxicology study in monkeys.
- IDX375 is a promising clinical candidate for the treatment of HCV infection and is undergoing IND-enabling studies.

References

1. Wasley A and Alter MJ (2000). Semin. Liver Dis. 20:1-16.
2. Cretton-Scott E, *et al* (2008). J Hepatology 48, S220.
3. Standring D, Lanford R, Cretton-Scott E, *et al* (2008). J Hepatology 48, S30.

Acknowledgments

The authors would like to thank the Idenix chemistry teams for their efforts in discovering and providing the IDX375 used for these studies. The authors also thank Teresa Dahlman for assistance with preparation of the poster.

Presented at the 44th Annual Meeting of the European Association for the Study of the Liver (EASL); Copenhagen, Denmark; April 22 – 26, 2009

Disclosures: All authors are current employees of Idenix Pharmaceuticals, Inc.

Idenix PHARMACEUTICALS